VANCE: first in human phase I study of a novel ChAdOx1-MVA 5T4 vaccine in low and intermediate risk prostate cancer

THE JENNER INSTITUTE
DEVELOPING INNOVATIVE VACCINES

I. Redchenko[1], F. Cappuccini[1], E. Pollock[1], R. Bryant[1], L. Carter[1], C Verrill[1], J. Hollidge[1], L. Goodwin[2], R. Harrop[3], P. Romero[4], S. Vigano[4], T. Evans[5], J. Catto[2], F. Hamdy[1], A.V.S. Hill[1]

[1]University of Oxford, Oxford, UK, [2]Royal Hallamshire Hospital, Sheffield, UK, [3]Oxford BioMedica plc, Oxford, UK, [4]Lausanne University Hospital, Lausanne, Switzerland, [5]Vaccitech Ltd, Oxford, UK

Study Design and Objectives

We evaluated a novel vaccination platform based on two replication-deficient viruses, chimpanzee adenovirus ChAdOx1 and MVA, targeting the oncofoetal antigen 5T4 in early stage prostate cancer patients. The study arms are shown on the right.

Primary objectives: - Safety and immunogenicity (*ex vivo IFNγ ELISPOT)*

Secondary objectives: - Tumour immune infiltration into the prostate (IHC of FFPE tissue, flow cytometry of fresh tissue)
- Serum PSA level change secondary to vaccination

Exploratory objectives: - Phenotype and functional profile of PBMCs
- T cell epitope mapping

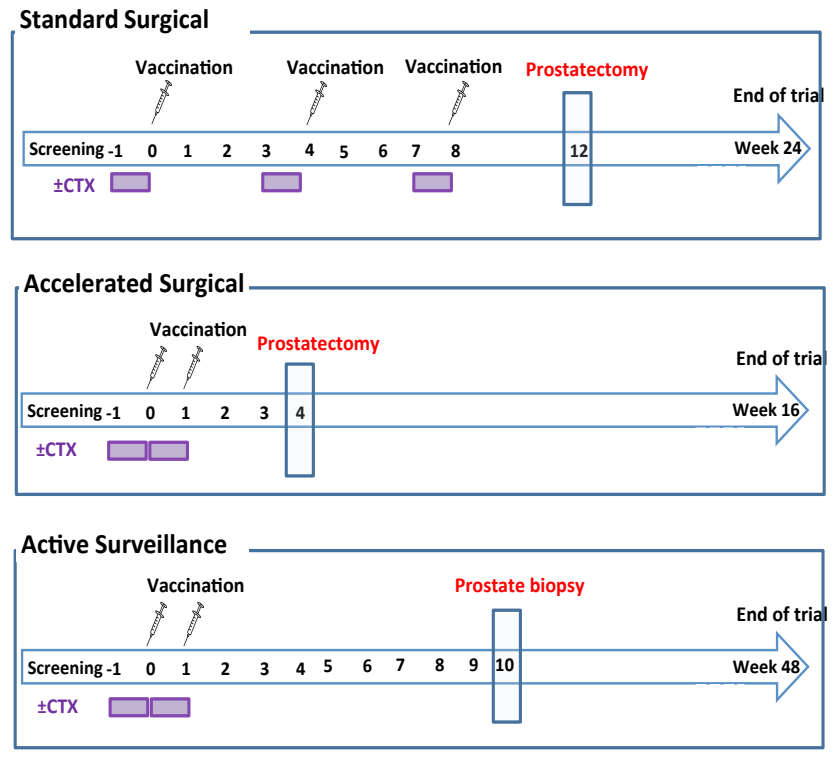

Standard Surgical
Vaccination
Vaccination
Vaccination
Prostatectomy
End of trial
Screening -1 0 1 2 3 4 5 6 7 8 12 Week 24
±CTX
Accelerated Surgical
Vaccination
Prostatectomy
End of trial
Screening -1 0 1 2 3 4 Week 16
±CTX
Active Surveillance
Vaccination
Prostate biopsy
End of trial
Screening -1 0 1 2 3 4 5 6 7 8 9 10 Week 48
±CTX

Conclusions

- *Both ChAd and MVA vaccines were well tolerated in all subjects*
- *Clear evidence of breaking tolerance*
 - Ex vivo 5T4-specific CD8 and CD4 T cell responses were induced in the majority of vaccinees
 - 5T4-specific CD4 and CD8 T cells were expanded from fresh prostate tissue of immunised subjects
 - CD8 T cell infiltration was detected in the FFPE surgical specimens of the vaccinated patients
- *Intriguing transient increase in PSA in the majority of the vaccinated patients*

Study Results

Vaccination induces ex vivo 5T4 T cell response in the majority of patients

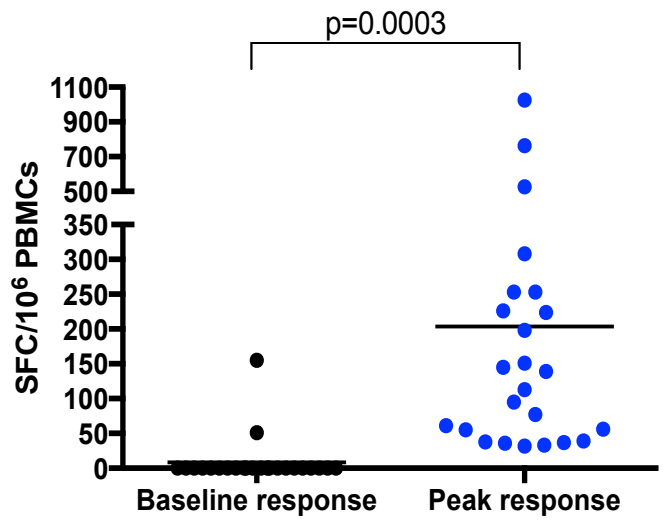

p=0.0003
SFC/10^6 PBMCs
1100 900 700 500 350 300 250 200 150 100 50 0
Baseline response
Peak response

5T4-specific T-cell responses to vaccination measured by IFNγ ELISPOT. Peak response, expressed as a number of the antigen-specific T cells secreting IFNγ per one million of PBMCs, in each patient who mounted the 5T4-specific T cells response following vaccination was compared to the 5T4 response detected at baseline. Bars represent medians.

Vaccination induces T cell response to a broad range of 5T4 T cell epitopes

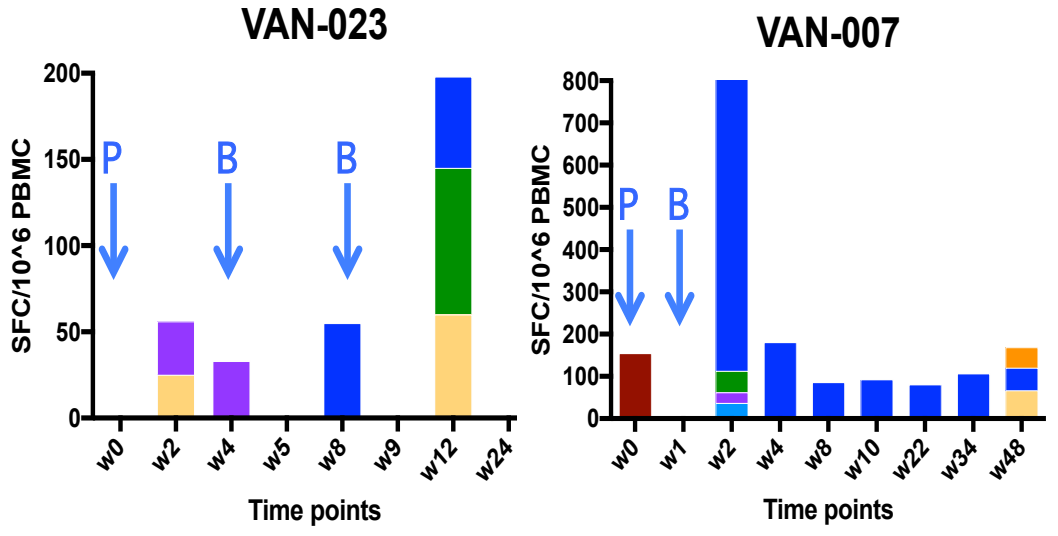

VAN-023
SFC/10^6 PBMC
P B B
w0 w2 w4 w5 w8 w9 w12 w24
Time points
VAN-007
SFC/10^6 PBMC
P B
w0 w1 w2 w4 w8 w10 w22 w34 w48
Time points

In the ELISPOT assay, the overlapping peptides spanning the entire 5T4 protein have been split into 8 pools (10 peptides per pool). The response to each individual pool is shown in a different colour. The magnitude and kinetics of the 5T4 T cell response are shown in 2 illustrative patients: one patient (VAN-023) was randomised to the standard vaccination regimen, another (VAN-007) – to the accelerated immunisation schedule. P indicates prime with ChAdOx.5T4; B indicates boost with MVA.5T4.

Kinetics of vaccine-induce T cell response

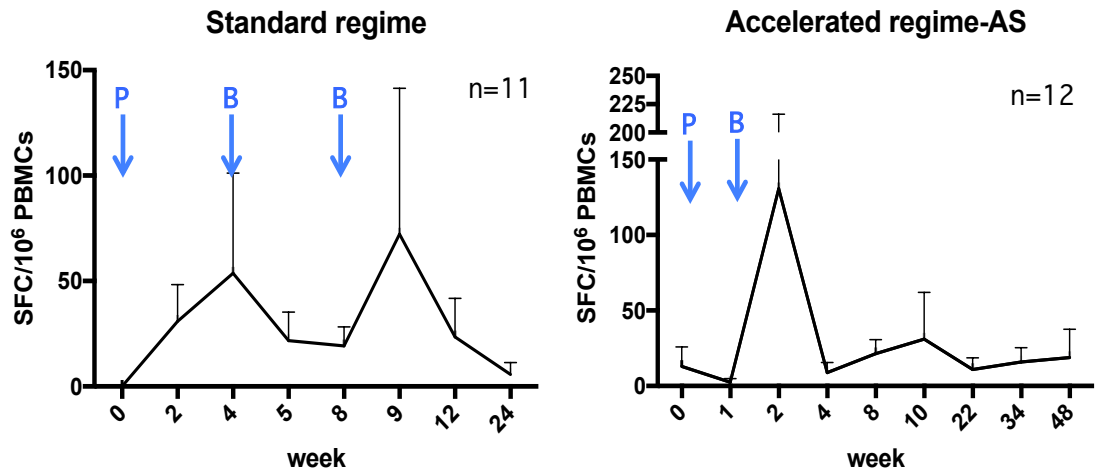

Standard regime
n=11
SFC/10^6 PBMCs
P B B
0 2 4 5 8 9 12 24
week
Accelerated regime-AS
n=12
SFC/10^6 PBMCs
P B T
0 1 2 4 8 10 22 34 48
week

5T4-specific T-cell responses to vaccination measured by IFNγ ELISPOT. Blood samples from vaccinated patients have been collected on the day of each vaccination, 7-14 days after each vaccination and at the follow-up visits up to week 48. The PBMCs have been exposed to the 5T4 peptide pools for 18 hours and and the numbers of the antigen-specific T cells secreting IFN-γ have been calculated per one million of PBMCs. The graphs represent an average number of 5T4-specific T cells at each timepoint in two study arms.

Vaccination induces transient increase in serum PSA

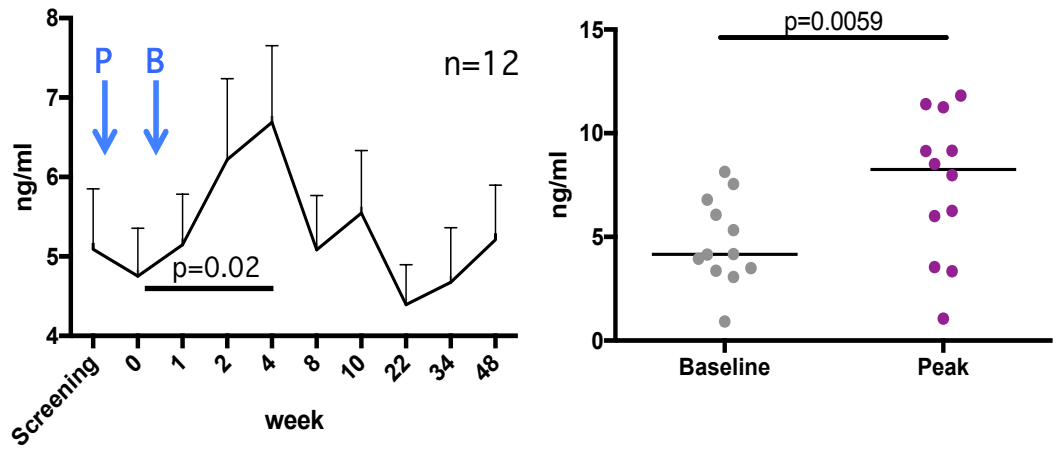

ng/ml
P B
n=12
p=0.02
Screening 0 1 2 4 8 10 22 34 48
week
p=0.0059
ng/ml
Baseline
Peak

A PSA level kinetics averaged for 12 vaccinated patients in the active surveillance (AS) arm during the study period is shown on the left. There is a statistically significant transient increase in PSA at week 4 compared to baseline. The graph on the right shows a maximum PSA level at any timepoint compared to baseline for each AS patient.

Illustrative patient VAN-010 results: ex vivo ELISPOT, flow cytometry of PBMCs and TILs, PSA kinetics and IHC

5T4-specific T cell response is detected ex vivo

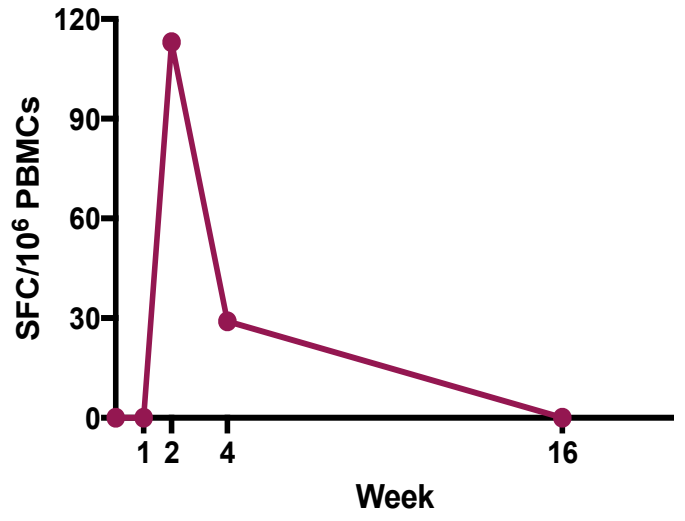

SFC/10^6 PBMCs
120 90 60 30 0
1 2 4 16
Week

VAN-010 was randomsied to the accelerated regimen to receive the ChAd vaccine at week 0, MVA at week 1 and to undergo surgery at week 4. Blood samples were collected at each clinic visit for IFNγ ELISPOT assay. The 5T4-specific T cell response peaked one week after the boosting MVA.5T4 immunisation.

5T4-specific CD8 T cells secreting IFN-γ and TNF-α are expanded from the blood and prostate tissue

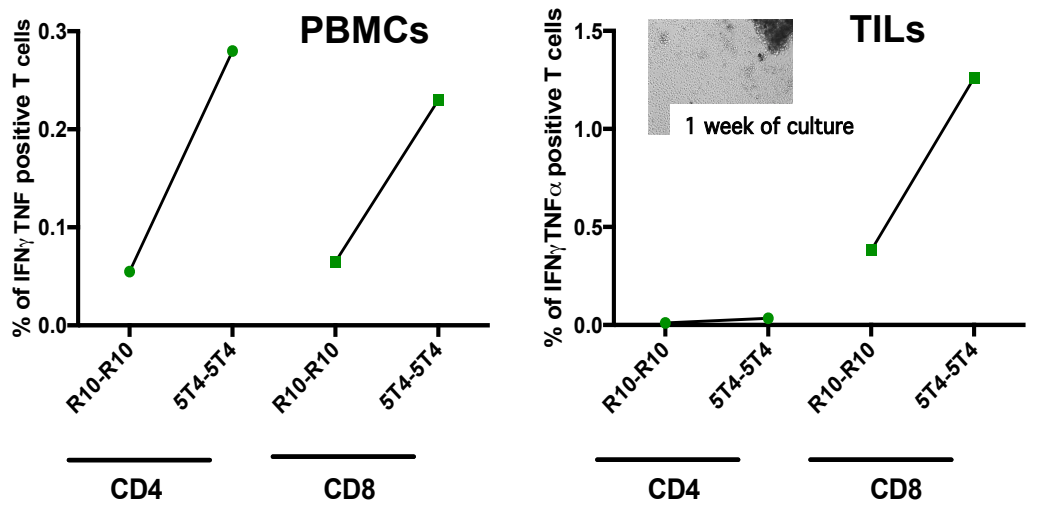

PBMCs
% of IFNγ TNF positive T cells
R10-R10 5T4-5T4 R10-R10 5T4-5T4
CD4 CD8
TILs
1 week of culture
% of IFNγ TNFα positive T cells
R10-R10 5T4-5T4 R10-R10 5T4-5T4
CD4 CD8

PBMCs and prostate surgical specimens were cultured either in medium alone or in the presence of the total 5T4 peptide pool to expand the relatively infrequent 5T4-specific T cells for further analysis by cytokine flow cytometry. ~0.2% of CD8 T cells in the blood and ~1% in the prostate secreted IFNγ and TNFα in response to 5T4 peptide pool stimulation. Both samples were collected at week 4 on the study. R10-R10 denotes the 5T4 antigen naïve T cells; 5T4-5T4 denotes the T cells expanded in the presence of 5T4 peptide pool.

Level of serum PSA sub-forms is increased following vaccination

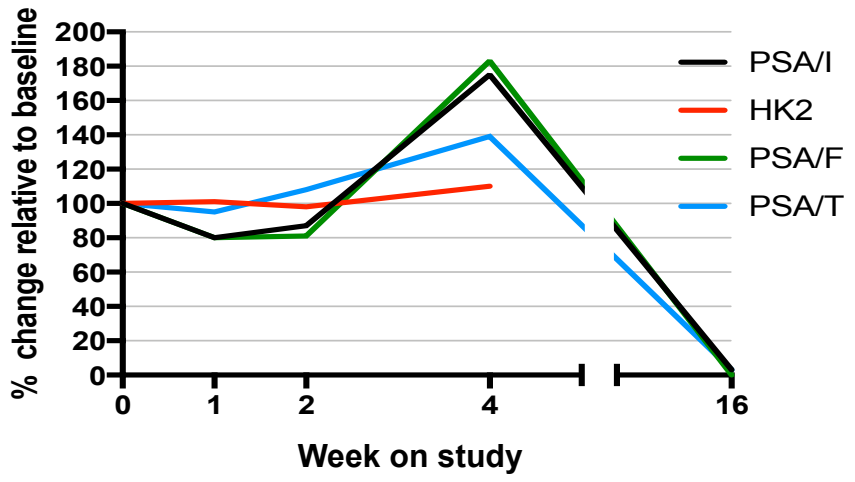

% change relative to baseline
0 1 2 4 16
Week on study
PSA/I
HK2
PSA/F
PSA/T

Free PSA (PSA/F) and intact PSA (PSA/I) have more rapid elimination kinetics compared to the total PSA (PSA/T). Therefore, these sub-forms could be a more sensitive readout of the vaccine effect on the target organ. As shown on the graph, there was an 80% increase over the baseline in the level of both intact and free PSA sub-forms following vaccination.

Immune cell infiltration including CD8 T cells is detected in prostate tissue by IHC

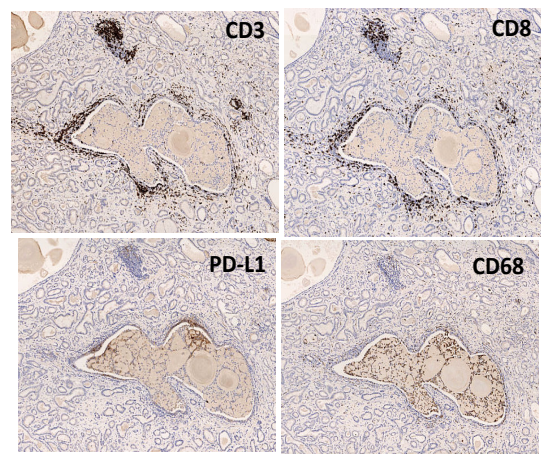

CD3
CD8
PD-L1
CD68

Consecutive sections of the FFPE surgical specimen were stained for CD3, CD8, CD68 and PD-L1 expression. The staining of paired pre-treatment biopsy samples is underway to quantify densities of immune cell subsets by digital image analysis (to be performed by Definiens).

ADVANCE trial of ChAdOx1-MVA 5T4 vaccine in combination with Nivolumab will commence in the summer 2018

Presented by Irina Redchenko, *irina.redchenko@ndm.ox.ac.uk*

The study was funded by the European Union Seventh Framework Programme under grant Agreement No. 602705 (Project IMPROVE)